Development and validation of a pharmacodynamic (PD) assay for TOS-358, the first covalent inhibitor of PI3Kα in clinical development

Mak, R., Bradley, J., Simpson, A., May, C., Wei, L., Ouellette, K., Olbrot, M., Blair, J., Chen, W., Dhawan, N, MacDougall, J.,
Totus Medicines, Emeryville, CA

Abstract

TOS-358 is a first-in-class covalent inhibitor of PI3Kα and is currently in clinical development in a variety of solid malignancies. We have developed a fit-for-purpose pharmacodynamic assay to evaluate the target occupancy of TOS-358 in both in vitro and in vivo settings at multiple timepoints. The PI3Kα assay allows direct measurement of both occupied and total protein in multiple different samples and tissue types. Signals of occupied and unoccupied PI3Kα proteins can be readily measured in cellular and in vivo samples. The assay was utilized in a large panel of cancer cell lines, revealing a direct correlation between target occupancy and pathway inhibition of markers such as phosphoAKT and phosphoS6. Critically, these correlations revealed the need to achieve near complete suppression of active PI3Kα to enable sustained pathway inhibition and cancer cell death. We furthermore utilized this assay to explore the turnover rate of PI3Kα across multiple diverse tumor types, mutations, and tissues. This approach enables highly sensitive target engagement analysis of TOS-358 across multiple formats, including in early clinical trials.

Assay Goals

The goals for the development of this assay are two-fold:

- Establish a method by which the relationship between dose and target occupancy can be explored in clinical samples. These data will help inform a recommended phase II dose and accelerate clinical development.
- Explore further the relationship between target occupancy and downstream effects on cancer cells

Assay Overview

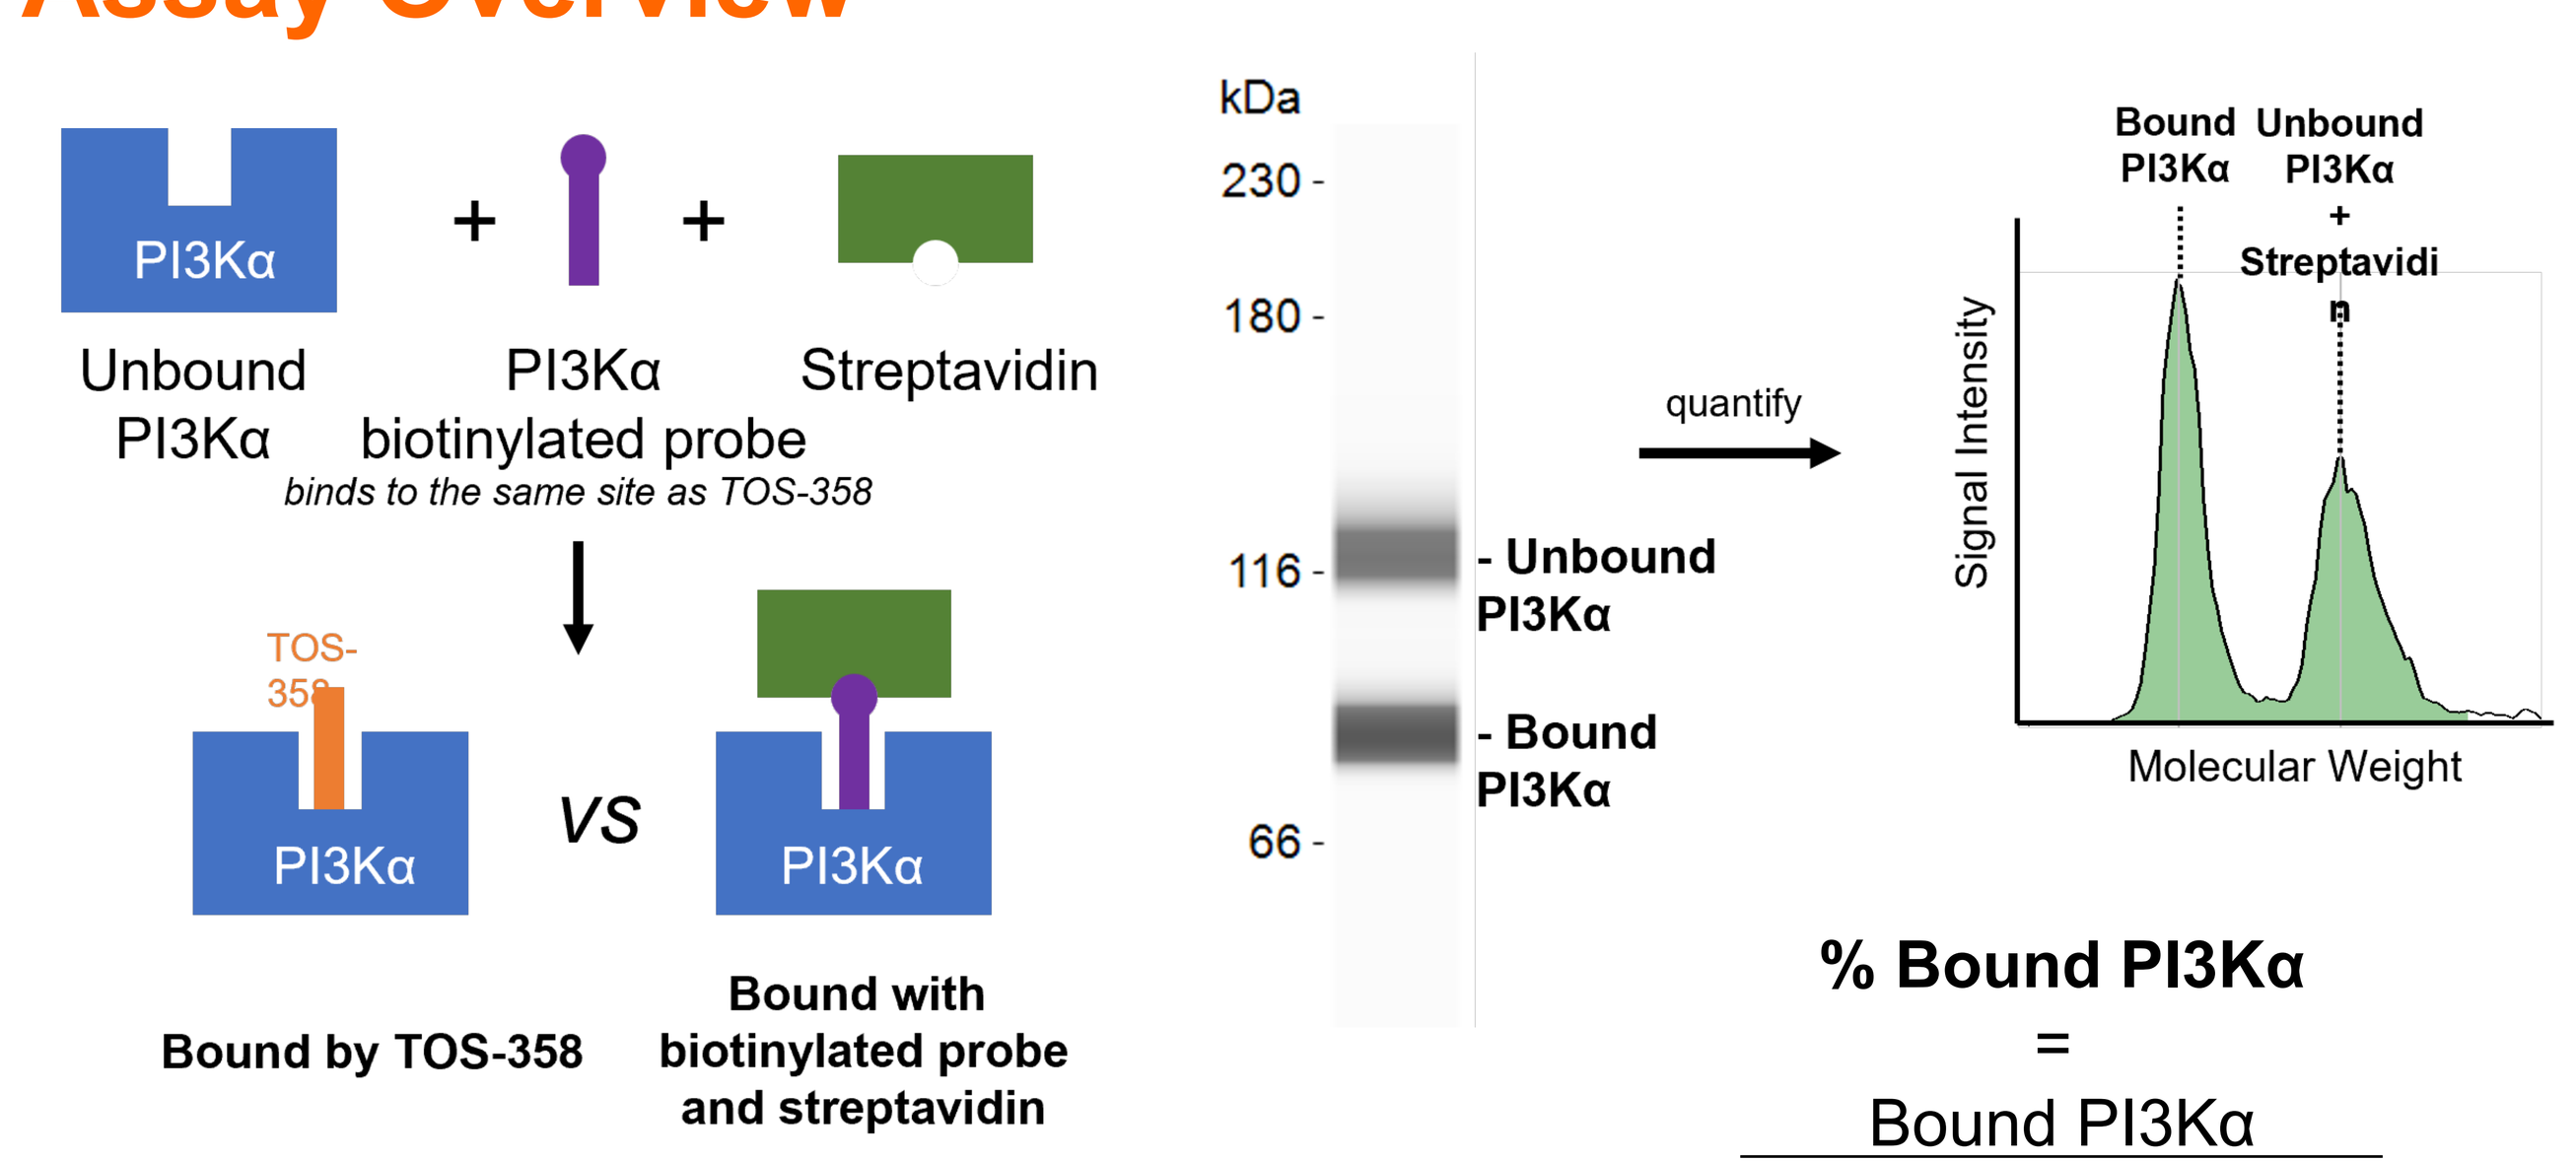
PI3Kα
Unbound PI3Kα
PI3Kα biotinylated probe
binds to the same site as TOS-358
Streptavidin
TOS-358
PI3Kα
vs
Bound by TOS-358
Bound with biotinylated probe and streptavidin
kDa
230
180
116
66
- Unbound PI3Kα
- Bound PI3Kα
quantify
Signal Intensity
Molecular Weight
Bound PI3Kα
Unbound PI3Kα + Streptavidin

% Bound PI3Kα = Bound PI3Kα / Total PI3Kα (bound + unbound)

Assay Description

The TOS-358 occupancy assay described herein is based on the Streptavidin Mass Shift Assay (SMaSh) proposed by Labenski et al (1). Briefly, cells exposed to TOS-358 will have a concentration dependent occupancy of PI3Kα protein within the cell leading to PI3Kα protein molecules occupied by TOS-358. Following TOS-358 exposure and cell lysis, the remaining PI3Kα molecules *not* occupied by TOS-358 are subsequently occupied with a biotinylated probe (TOS-258) through treatment of cell lysates. The biotinylated probe provides a basis for mass addition through the high affinity binding of streptavidin. Electrophoretic based protein separation coupled with antibody based PI3Kα detection allows for the quantification of TOS-358 bound (i.e. unable to bind probe) versus TOS-358 unbound (i.e. able to bind probe) PI3Kα in the sample. This strategy has been used in the development of covalent BTK and KRAS inhibitors (2-4) and has successfully been used to accelerate clinical development by guiding RP2D selection.

Results

TOS-358 displays a highly unique profile as the only clinical covalent inhibitor of PI3Kα

	PI3Kα-mutant IC_{95}	Average Cellular pAKT inhibition (pAKT/tAKT, n=7, 1uM)	Cellular Anti-Proliferative EC90 (n=10)	Predicted C_{trough} Requirement[d]	PI3Kβ Binding IC_{50}	PI3Kα IC_{95}/ PI3Kβ IC_{50} Ratio	Glucose Uptake Impact at Cellular EC90[e]
TOS-358	120nM[a]	96%	251nM	0nM[d]	2418nM	20X	10%
Alpelisib	3857nM[a]	85%	3585nM	3857nM[a]	10008nM	3X	84%
Pan-Mut Specific ($PI3K\alpha^{PAN}$ inh)	1015nM[b]	61%	6490nM	1015nM[b]	11798nM	10X	72%
H1047X-Mut Specific ($PI3K\alpha^{H1047R}$ inh)	8701nM[b]	-[c]	-[c]	8701nM[b]	-[c]	-[c]	43%

a-IC_{95} measured using the ATP-competitive NanoBRET assay
b-IC_{95} measured using Nanosyn kinase assay
c-Not measured as molecule can only impact H1047R cell lines and no PI3Kβ activity was measured. 70% of PI3K
d- Predicted C_{trough} minimum concentration in patients for single agent activity and retained PI3Kα IC_{95}. TOS-358 achieves continuous inhibition in absence of drug.
e- Zenbio assay was performed per standard protocol (link).

Isolated PBMCs show concentration dependent TOS-358 occupancy

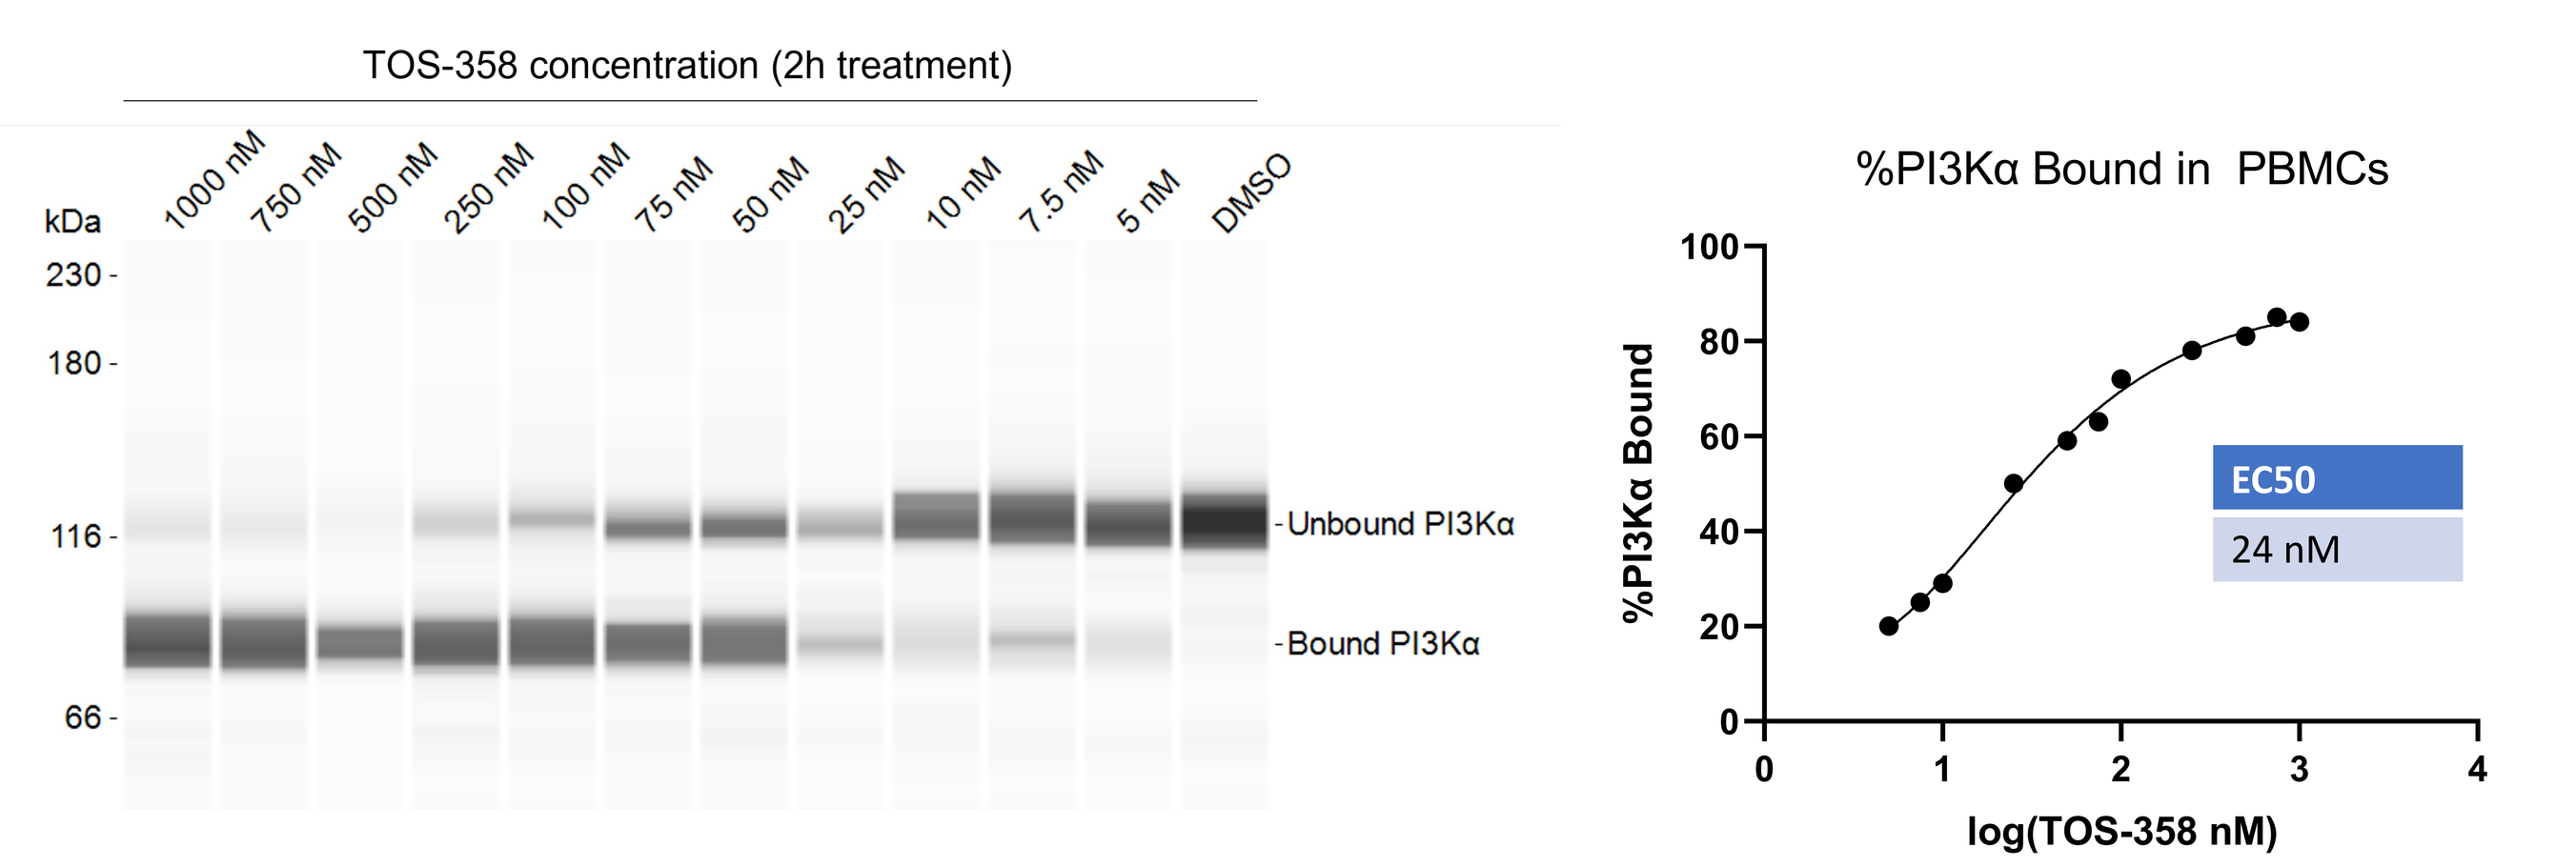
TOS-358 concentration (2h treatment)
1000 nM
750 nM
500 nM
250 nM
100 nM
75 nM
50 nM
25 nM
10 nM
7.5 nM
5 nM
DMSO
kDa
230
180
116
66
Unbound PI3Kα
Bound PI3Kα
%PI3Kα Bound in PBMCs
%PI3Kα Bound
log(TOS-358 nM)
EC50
24 nM

Purified PBMCs were obtained and treated with TOS-358 in culture for 2 hours, followed by processing for TOS-358 occupancy. These cells show a concentration dependent % occupancy of PI3Kα. The EC50 for occupancy in PBMCs is 24 nM.

Proof of concept validation of TOS-358 occupancy assay in PBMCs isolated from treated whole blood

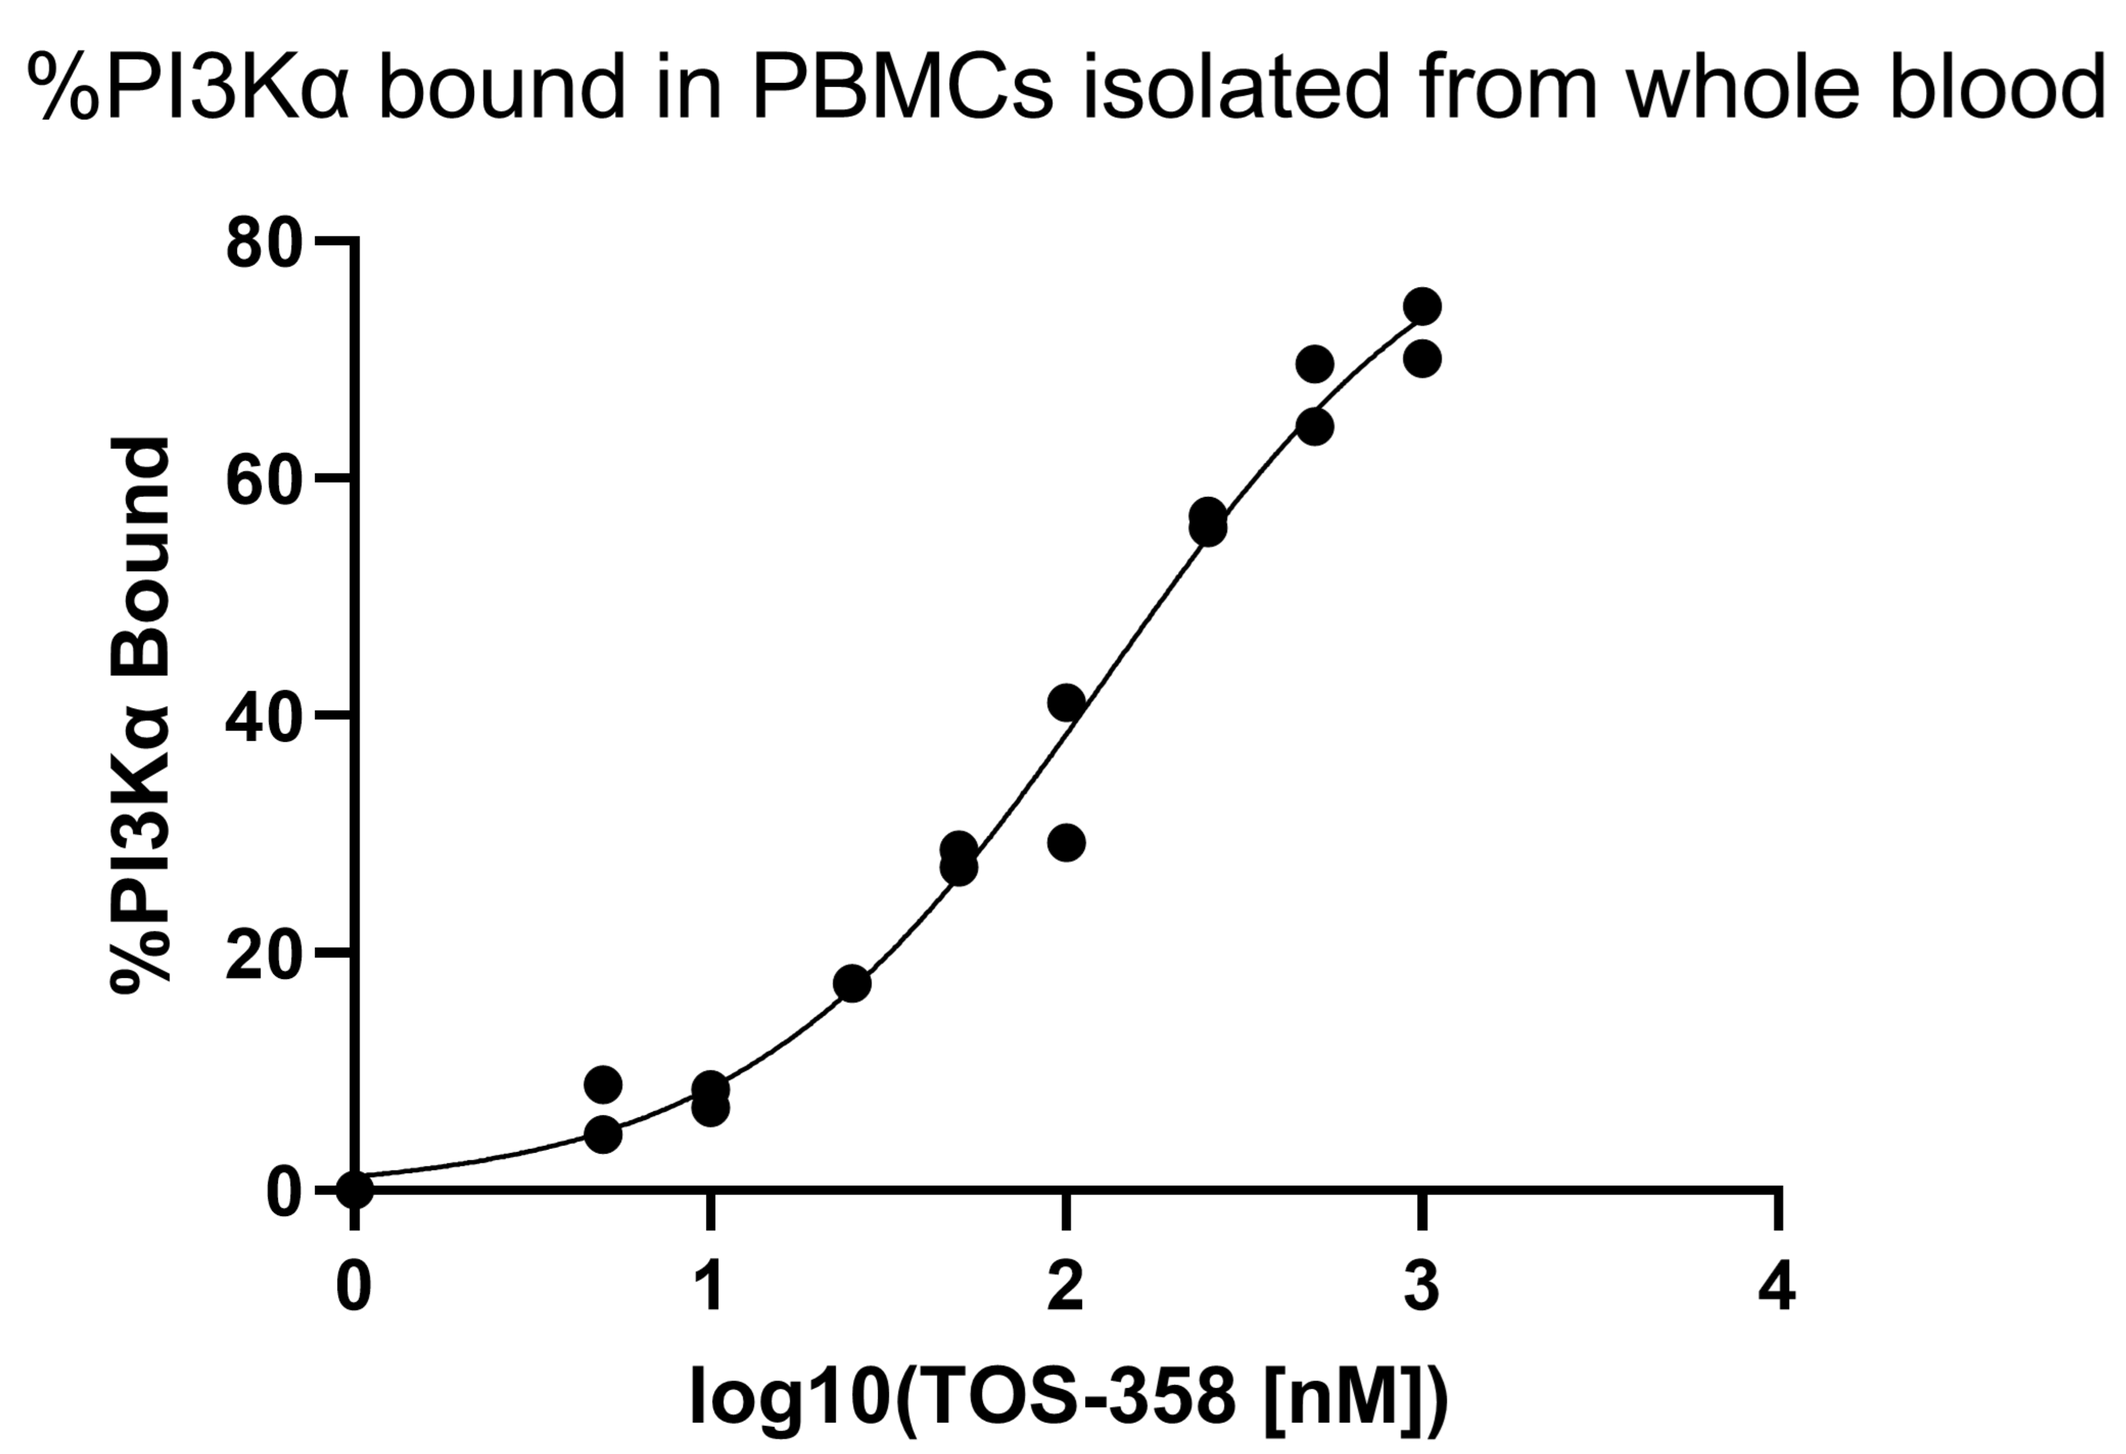
%PI3Kα bound in PBMCs isolated from whole blood
%PI3Kα Bound
log10(TOS-358 [nM])

The use of PBMCs as the source of clinical material was further validated using samples of whole blood. In this case, samples of whole blood were treated with different concentrations of TOS-358 for 2 hours followed by Ficoll gradient isolation of PBMCs, cell lysis, and analysis using the occupancy assay. This experiment revealed a TOS-358 concentration dependent occupancy in PBMCs from whole blood.

Occupancy is highly correlated with in-cell binding and proliferation inhibition in human breast cancer cells

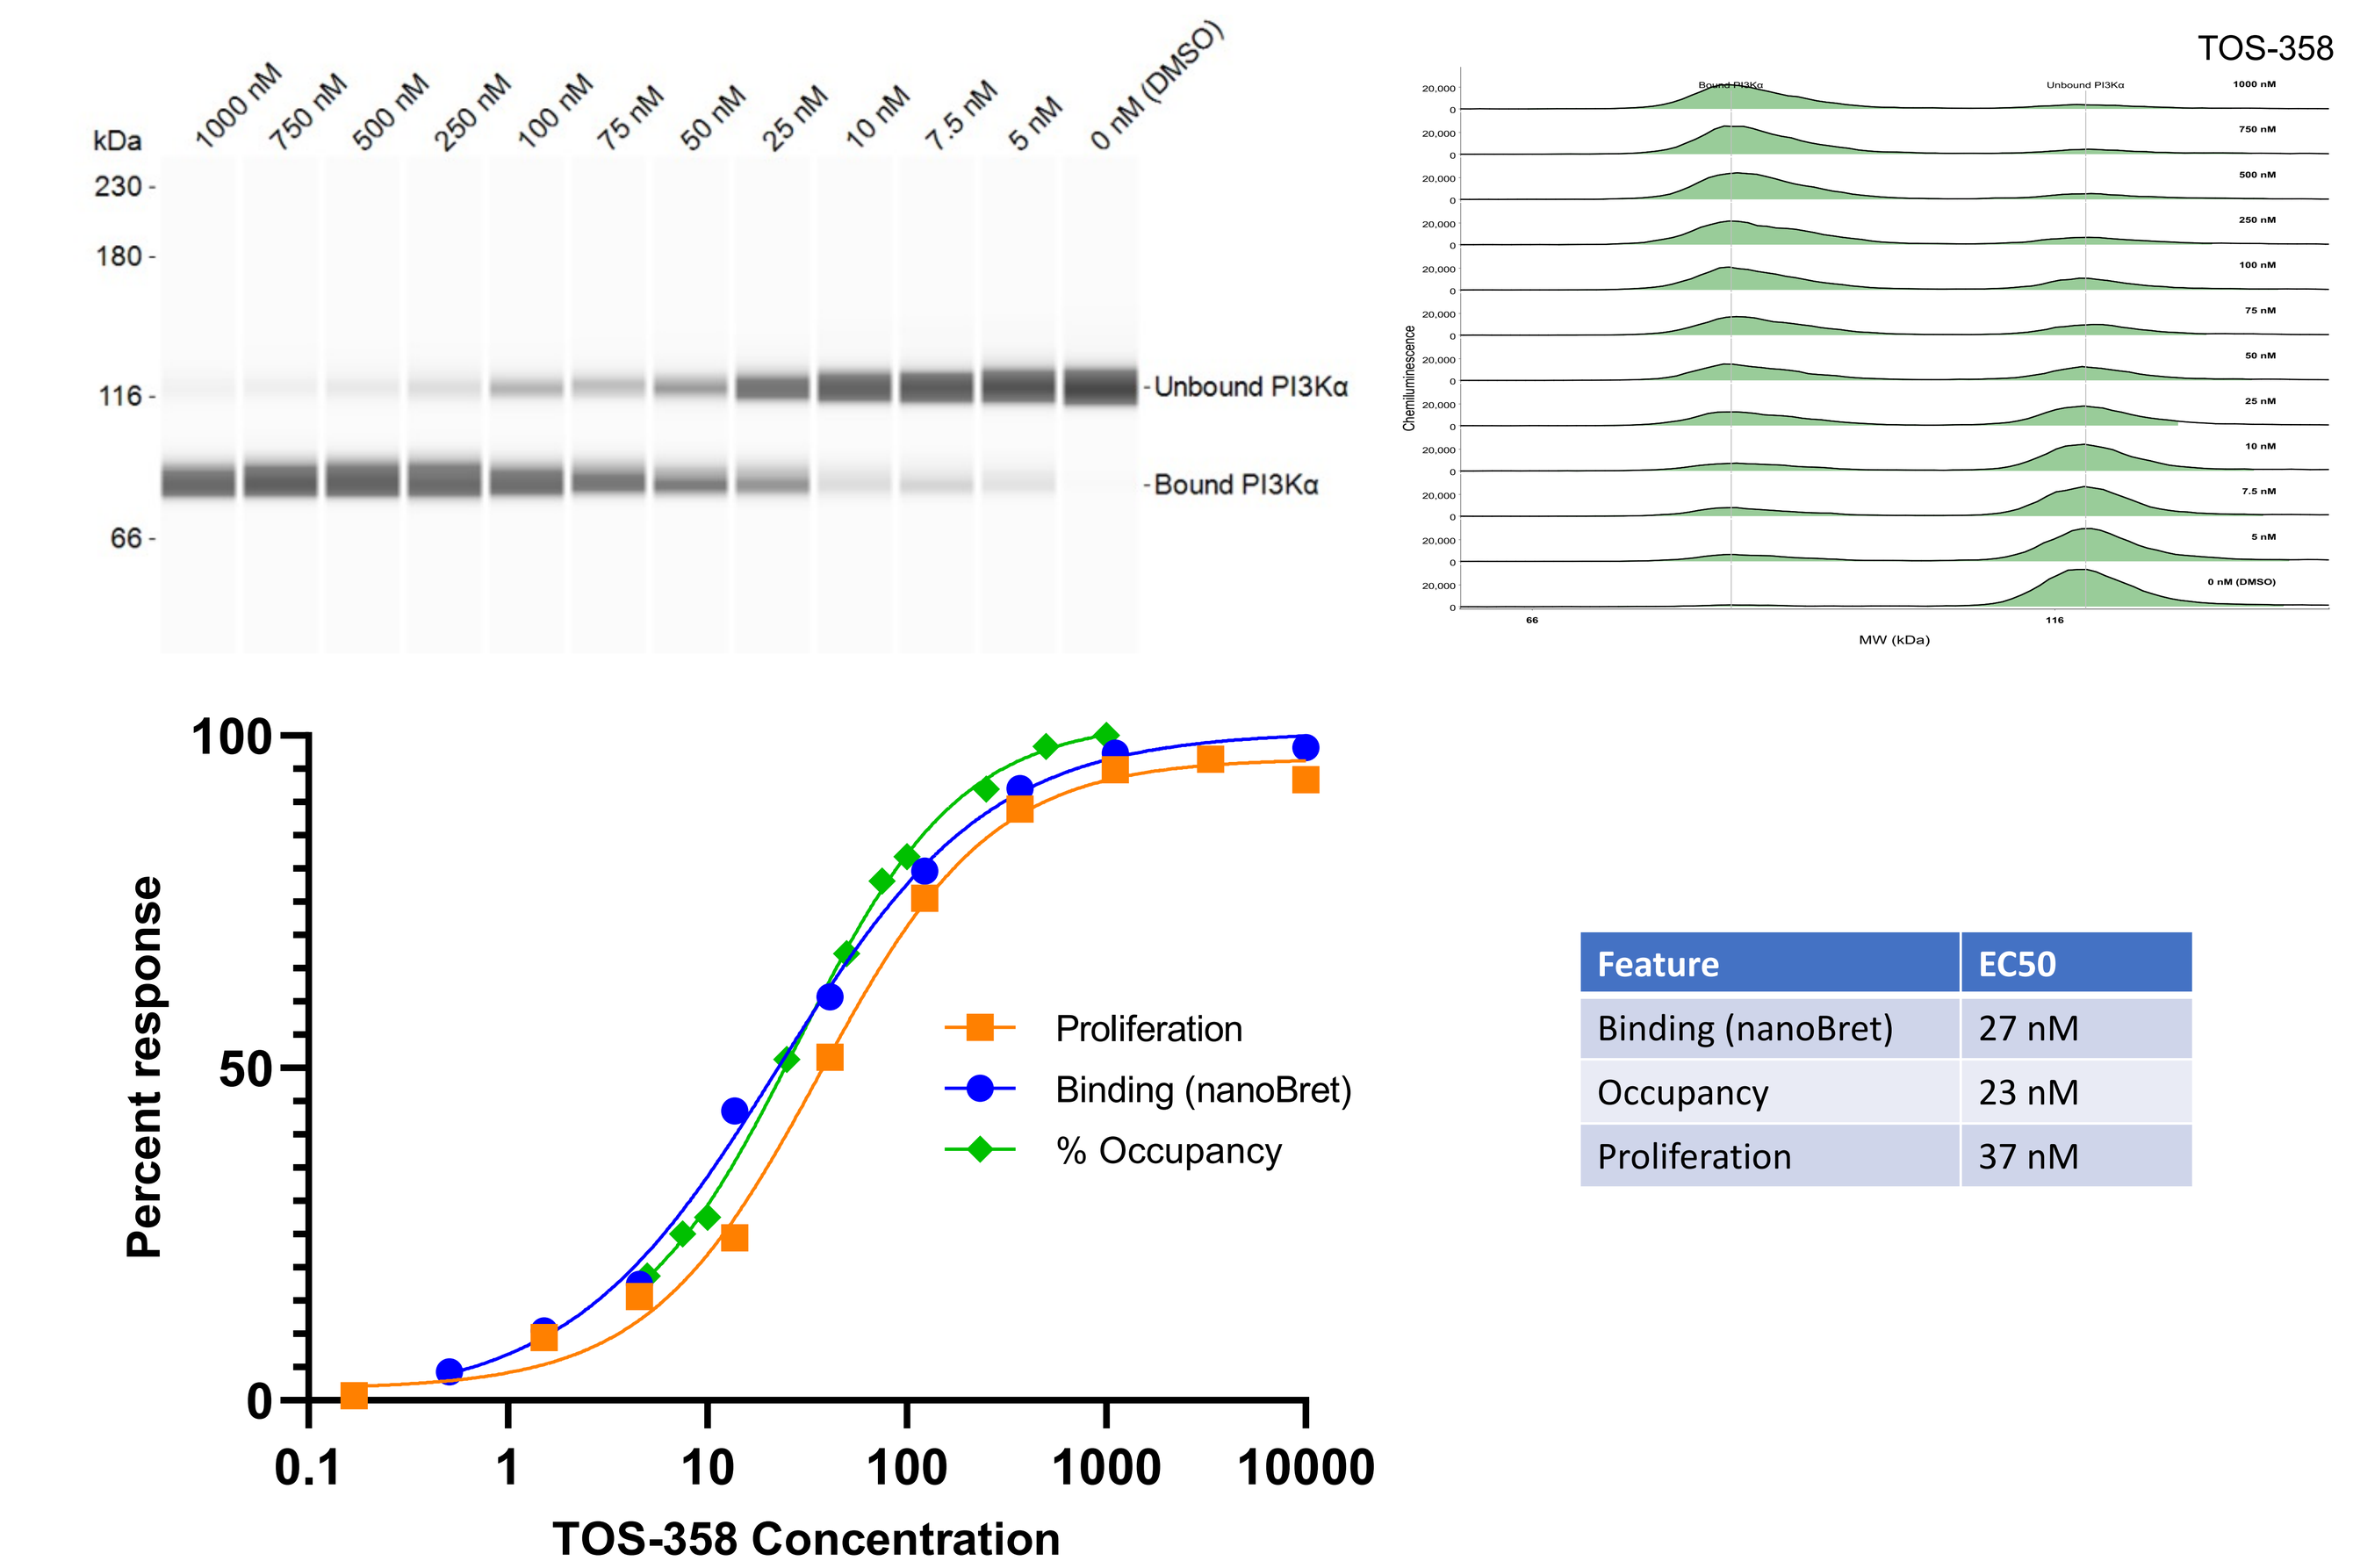
1000 nM
750 nM
500 nM
250 nM
100 nM
75 nM
50 nM
25 nM
10 nM
7.5 nM
5 nM
0 nM (DMSO)
kDa
230
180
116
66
Unbound PI3Kα
Bound PI3Kα
TOS-358
Percent response
TOS-358 Concentration
Proliferation
Binding (nanoBret)
% Occupancy
Feature | EC50
Binding (nanoBret) | 27 nM
Occupancy | 23 nM
Proliferation | 37 nM

The relationship between TOS-358 PI3Kα occupancy, in-cell PI3Kα binding, and proliferation was explored by estimating EC50s for each of these parameters in cells in culture. The data from this comparison revealed these measures to be closely related - with the EC50 values for % occupancy to be 27 nM, the EC50 for binding in the nanoBret assay to be 23 nM and the EC50 for inhibition of proliferation to be 37 nM in T47D cells.

Slow turnover highlights durable target inhibition by TOS-358 in cancer cells

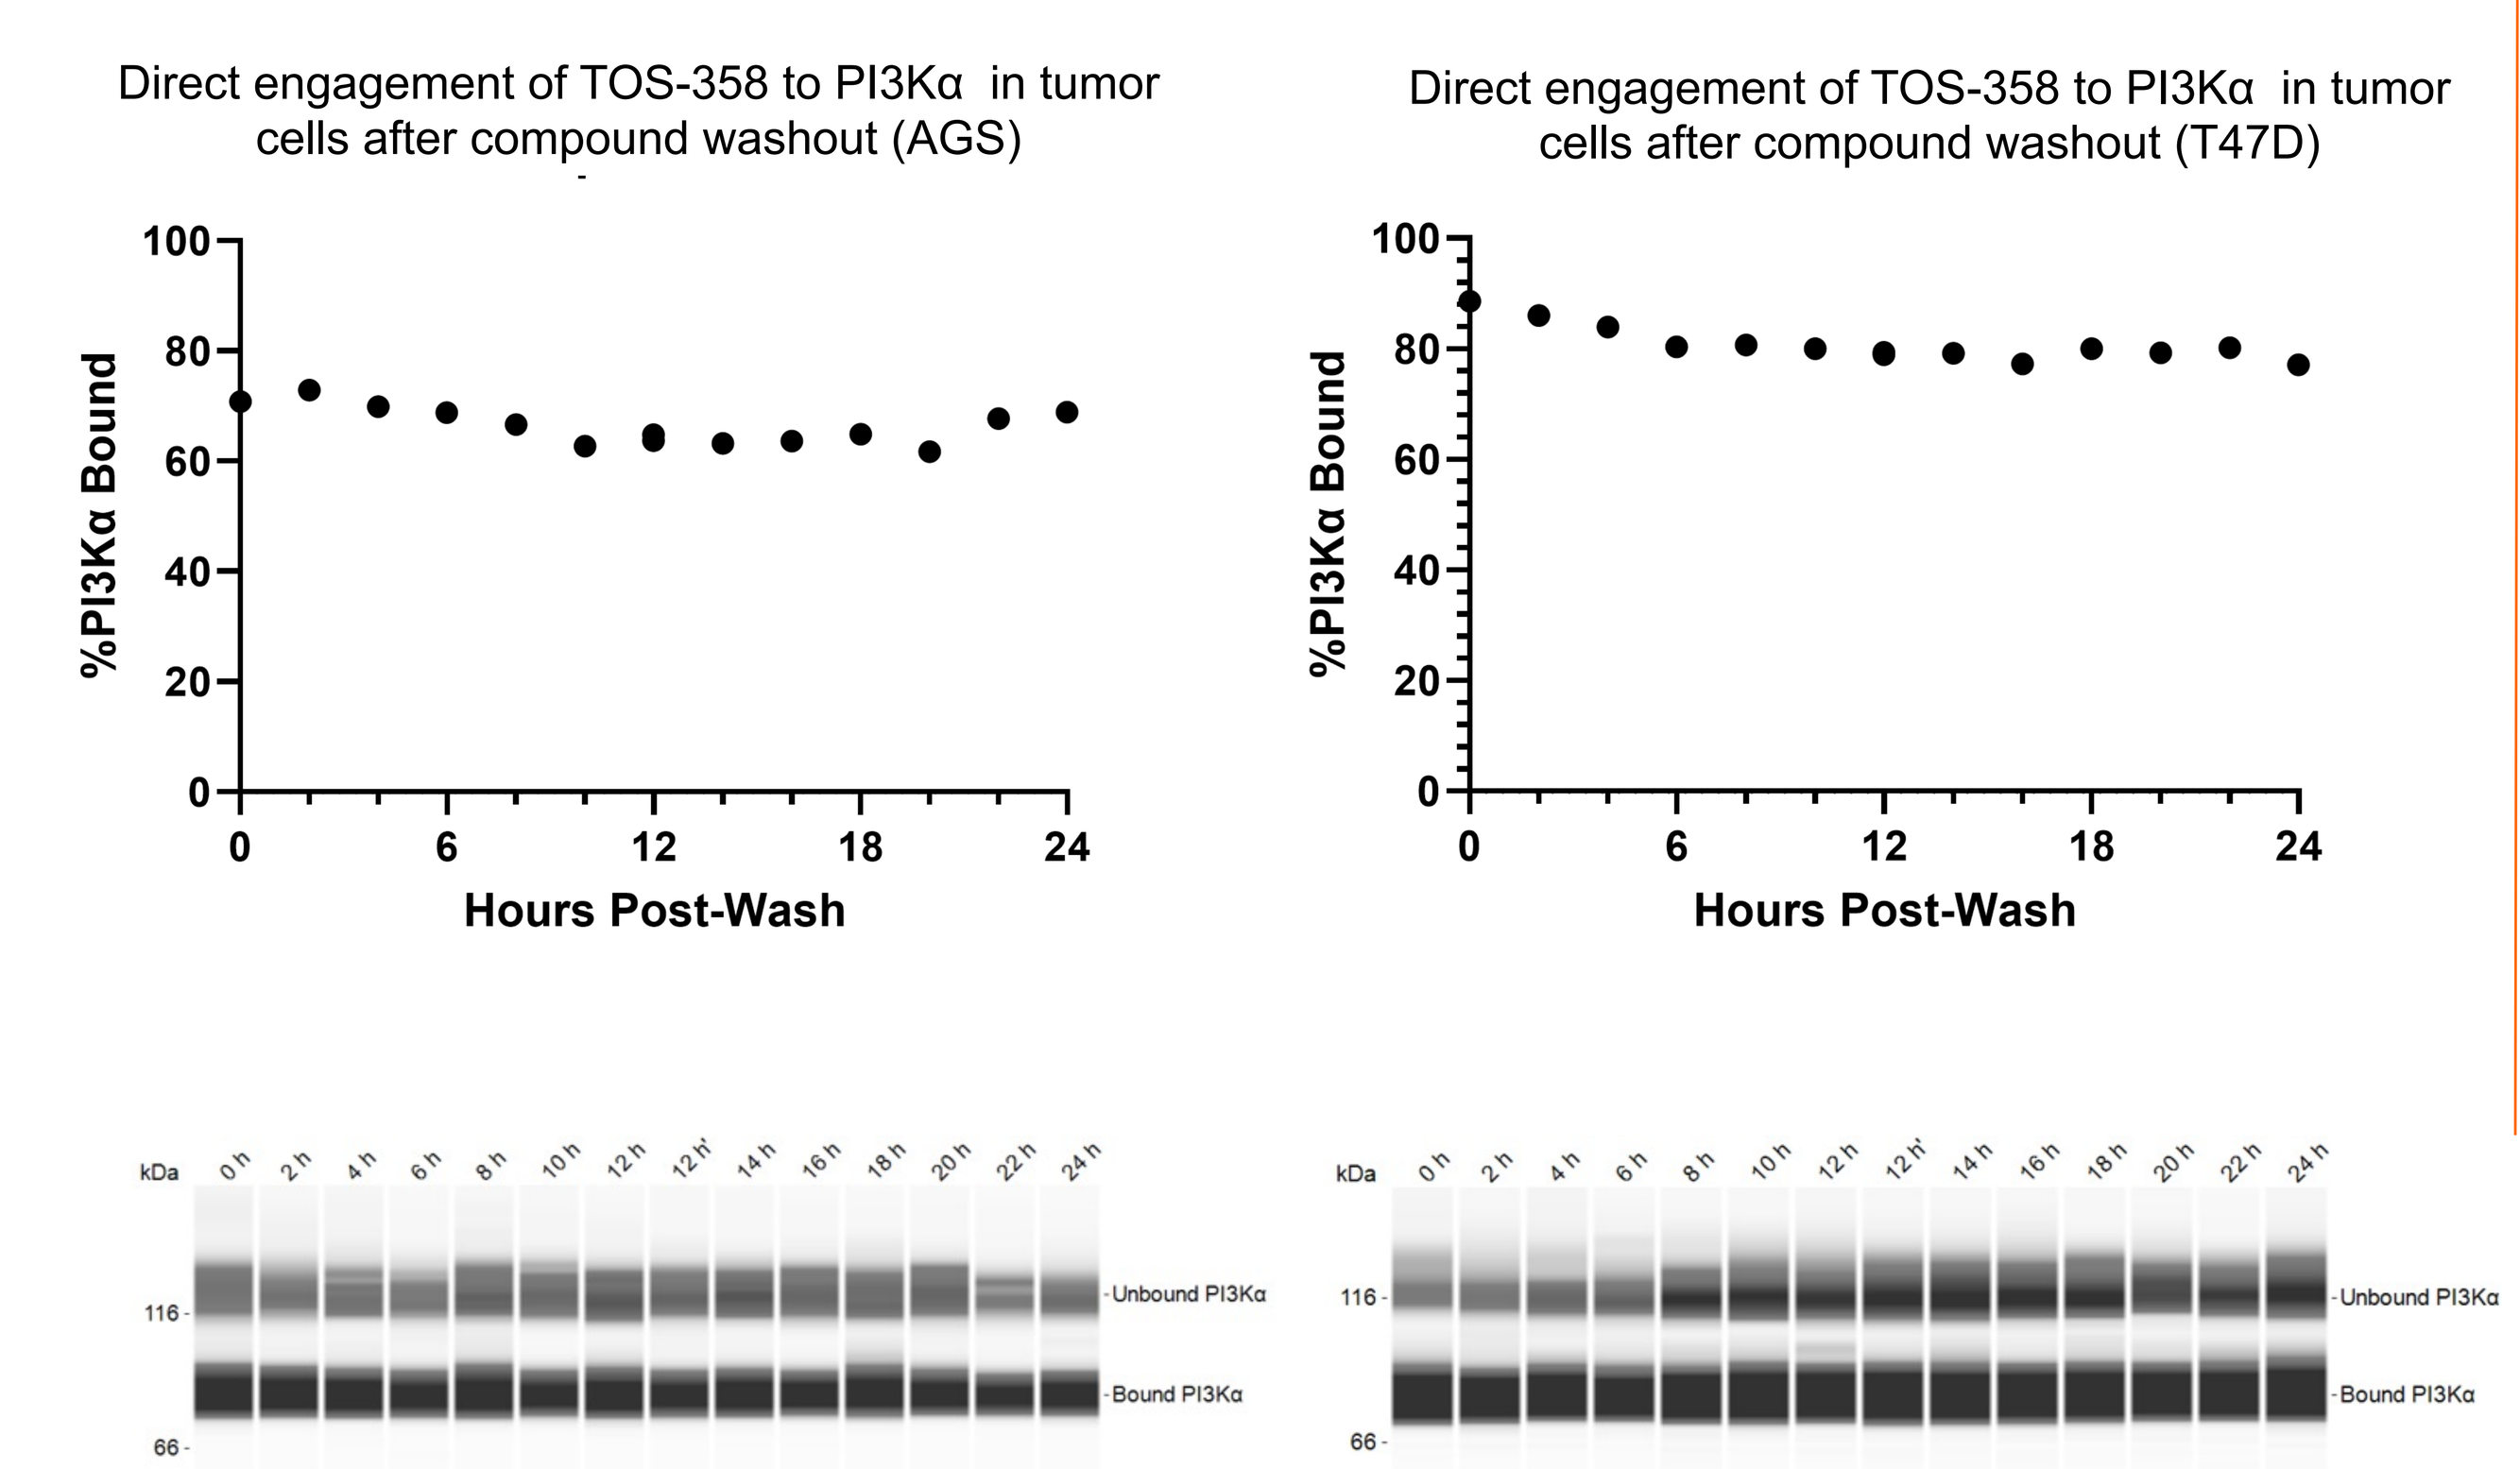
Direct engagement of TOS-358 to PI3Kα in tumor cells after compound washout (AGS)
Direct engagement of TOS-358 to PI3Kα in tumor cells after compound washout (T47D)
%PI3Kα Bound
Hours Post-Wash
Unbound PI3Kα
Bound PI3Kα

Given that TOS-358 is a covalent inhibitor of PI3Kα we utilized the occupancy assay to explore the PI3Kα protein turnover in cancer cell lines. We hypothesized that the degradation of TOS-358 bound PI3Kα could be monitored over time and would be reflected in a temporal change in % occupancy. The results of these experiments reveal that the turnover of PI3Kα in cancer cells is very slow, highlighting the durable target occupancy and pathway inhibition achievable by TOS-358. This results in durable efficacy upon low, pulsatile dosing of TOS-358 as has been observed in multiple cancer models across diverse PI3Kα-mutant histologies

TOS-358 occupancy correlates with inhibition of pathway signaling in cancer cell lines

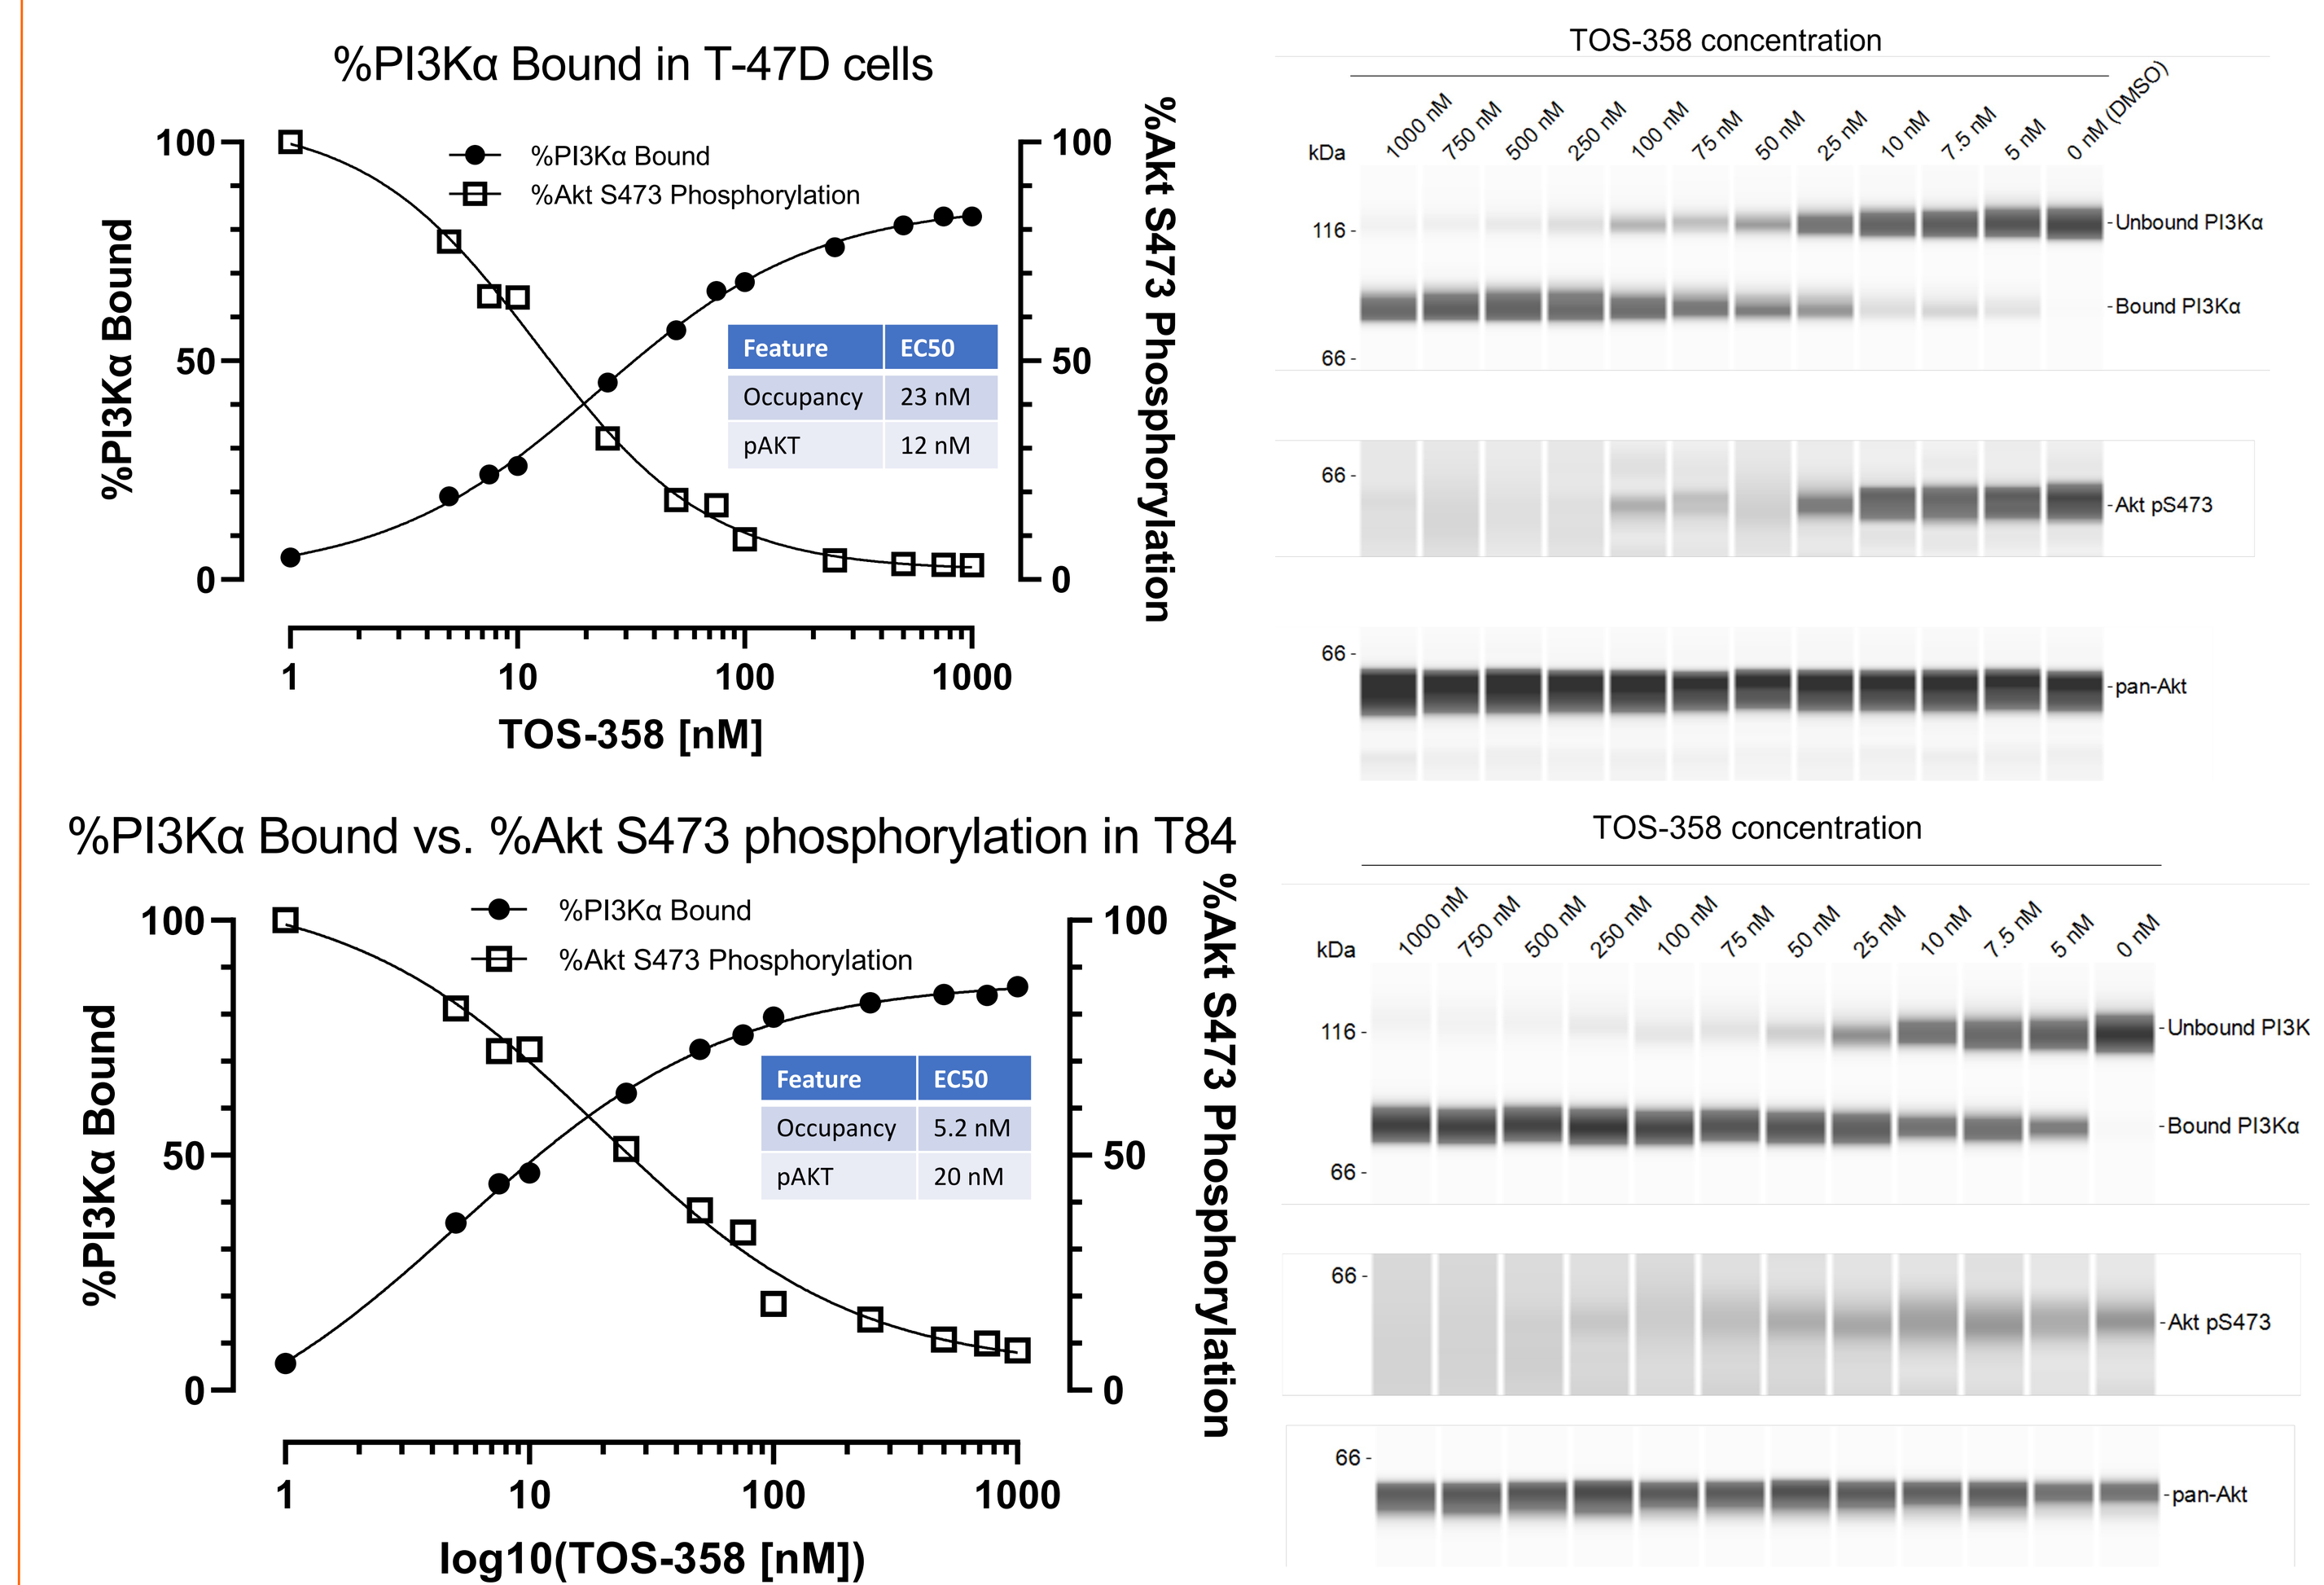
%PI3Kα Bound in T-47D cells
%PI3Kα Bound
%Akt S473 Phosphorylation
TOS-358 [nM]
Feature | EC50
Occupancy | 23 nM
pAKT | 12 nM
%PI3Kα Bound vs. %Akt S473 phosphorylation in T84
log10(TOS-358 [nM])
Feature | EC50
Occupancy | 5.2 nM
pAKT | 20 nM
TOS-358 concentration
Unbound PI3Kα
Bound PI3Kα
Akt pS473
pan-Akt

Cultured cells were treated with different concentrations of TOS-358 and then harvested for analysis. These experiments reveal that maximum occupancy of PI3Kα results in complete pathway inhibition as measured by pAKT (S473).

Saturation of target occupancy suggests cells have a small population of inactive PI3Kα

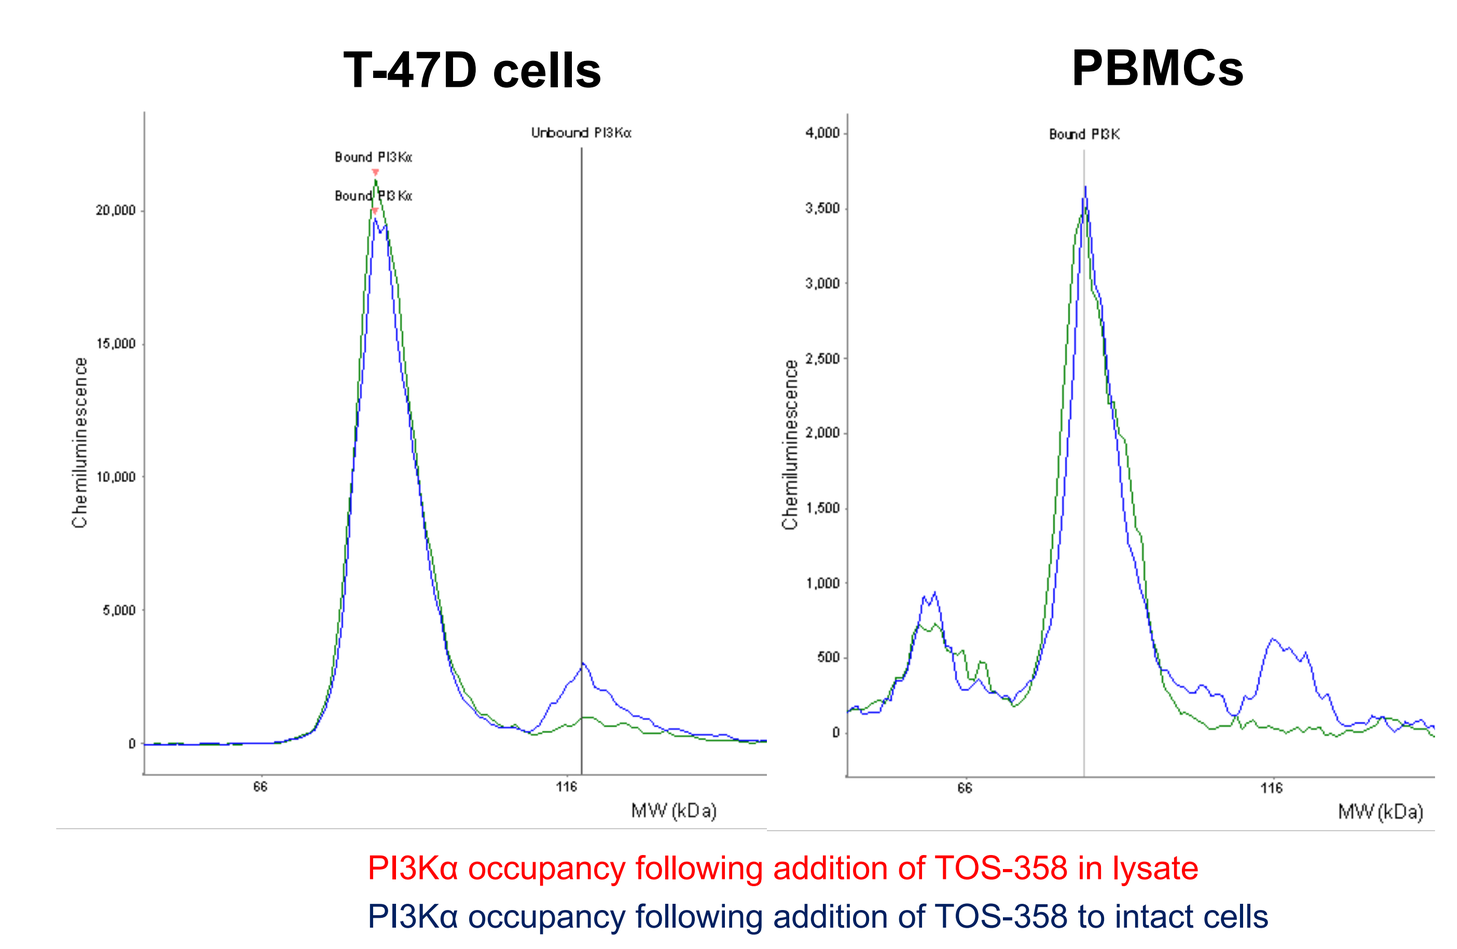
T-47D cells
PBMCs
Chemiluminescence
MW (kDa)
PI3Kα occupancy following addition of TOS-358 in lysate
PI3Kα occupancy following addition of TOS-358 to intact cells

We have observed that this assay saturates at ~70-90% of PI3Kα occupancy in a variety of different cell lines. However, at saturating doses, we see no remaining PI3Kα activity and no phosphorylation of downstream effectors such as AKT or PRAS40. This implies that a subset of PI3Kα in cells is both inactive and not accessible to drug. This is not unexpected as PI3Kα activation requires a complex set of interactions with multiple regulatory subunits.

Conclusions

- The covalent nature of TOS-358 allows for the assessment of target occupancy in a variety of sample types.
- Target occupancy is tightly correlated to in-cell binding, inhibition of pathway signaling, and proliferation.
- Cells of the B-cell lineage express PI3Kα and therefore allow for the measurement of target engagement using PBMCs as a tissue source.
- Assessment of PI3Kα occupancy in samples of whole blood will provide guidance on the effective therapeutic window and RP2D in clinical trials. TOS-358 is currently in a Phase 1 clinical trial.
- Please visit Poster #CT245 on Apr. 18, 2023, 1:30 PM - 5:00 PM for a presentation of our TOS-358 clinical trial.

References

1) Labenski MT, et al. SMaSh: A Streptavidin Mass Shift Assay for Rapidly Quantifying Target Occupancy by Irreversible Inhibitors. Biochemistry. 2021 Oct 5;60(39):2915-2924.
2) Advani RH, et al. Bruton tyrosine kinase inhibitor ibrutinib (PCI-32765) has significant activity in patients with relapsed/refractory B-cell malignancies. J Clin Oncol. 2013 Jan 1;31(1):88-94.
3) Sun C, et al. Clinical and biological implications of target occupancy in CLL treated with the BTK inhibitor acalabrutinib. Blood. 2020 Jul 2;136(1):93-105.
4) Hong DS, et al. KRASG12C Inhibition with Sotorasib in Advanced Solid Tumors. N Engl J Med. 2020 Sep 24;383(13):1207-1217.
5) https://servers.binf.ku.dk/bloodspot/?gene=PIK3CA&dataset=DMAP

Please visit www.totusmedicines.com for a PDF version of this poster